Poster # P1.02_12003

The Role of Lung Cancer Advocacy Organizations in Biomarker Testing

Win Boerckel, LCSW-R, MBA, MSW[1]; Carolyn Aldigé[2]; Upal Basu Roy, PhD, MPH[3]; Andrew Ciupek, PhD[4]; Dusty Donaldson, MA[5]; Hildy Grossman, PhD[6]; Cindy Langhorne[7]; Lauren S. Rosenthal, MPH[8,11]; Claire Saxton, MBA[9]; Gerard A. Silvestri, MD, MS[10,11]; Robert A. Smith, PhD[8,11]; Linda Wenger[3]

[1]CancerCare, Syosset, NY; [2]Prevent Cancer Foundation, Alexandria, VA; [3]LUNGevity Foundation, Bethesda, MD; [4]Lung Cancer Alliance, Washington, DC; [5]Dusty Joy Foundation (LiveLung), High Point, NC; [6]Upstage Lung Cancer, Brookline, MA; [7]Caring Ambassadors Lung Cancer Program, Lakewood, WA; [8]American Cancer Society, Atlanta, GA; [9]Cancer Support Community, Washington, DC; [10]Medical University of South Carolina, Charleston, SC; [11]National Lung Cancer Roundtable

Background

- Lung cancer is not just a single disease, but rather comprises many subtypes, each with unique genetic changes (or driver mutations) that allow the cancer to develop and grow (**Figure 1**)
- These unique mutations serve as biomarkers to:
 - Help doctors classify an individual's specific type of lung cancer
 - Help determine the most appropriate treatment[1]
- Most new treatment options available and under development for non-small cell lung cancer (NSCLC) are targeted therapies (target-specific biomarkers) or immunotherapies
- Clinical trials have shown significant improvement with targeted therapies in:[1-4]
 - Overall and progression-free survival
 - Health-related quality of life
- Despite this progress, biomarker testing is often viewed as an optional service by patients and presented as optional by their healthcare team

Figure 1. Driver Mutations in Lung Adenocarcinoma and Squamous Cell Lung Cancer*

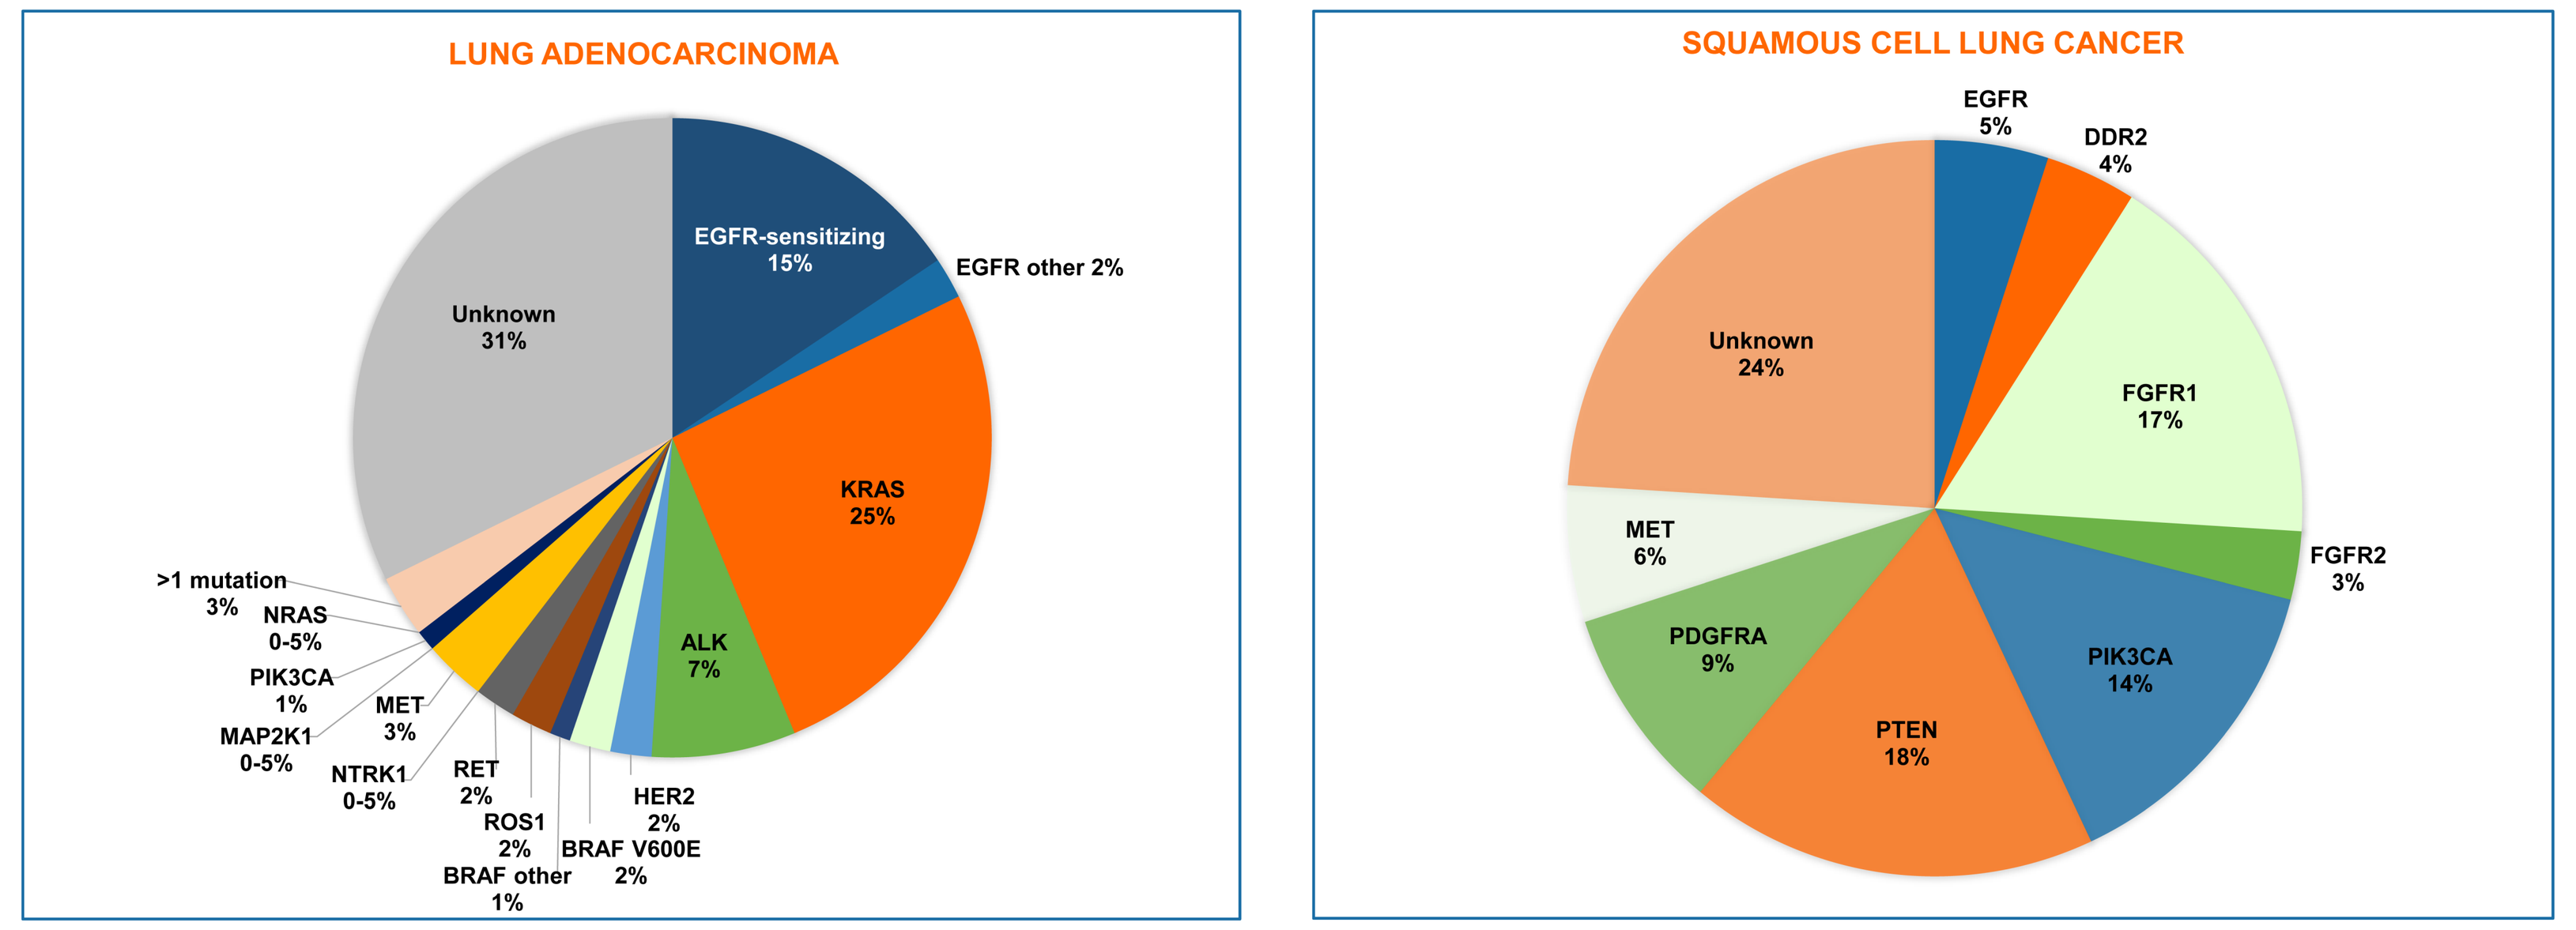
LUNG ADENOCARCINOMA
EGFR-sensitizing 15%
EGFR other 2%
KRAS 25%
ALK 7%
HER2 2%
BRAF V600E 2%
BRAF other 1%
ROS1 2%
RET 2%
NTRK1 0-5%
MAP2K1 0-5%
MET 3%
PIK3CA 1%
NRAS 0-5%
>1 mutation 3%
Unknown 31%
SQUAMOUS CELL LUNG CANCER
EGFR 5%
DDR2 4%
FGFR1 17%
FGFR2 3%
PIK3CA 14%
PTEN 18%
PDGFRA 9%
MET 6%
Unknown 24%

*Figure reused with permission from LUNGevity Foundation.

Objectives

- In March 2018, directors from lung cancer patient advocacy organizations and key opinion leaders in the lung cancer field held a roundtable to discuss trends in biomarker testing for patients with lung cancer. The objective of the roundtable was to align strategies to optimize patients' and physicians' awareness of biomarker testing to:
 - Increase uptake of biomarker testing
 - Ensure all lung cancer patients receive the most effective treatment

Findings

Barriers to Biomarker Testing

- Lack of patient and physician awareness
- Lack of unified language to describe testing
- Inadequate tissue sampling
- Slow turn-around time
- Limited availability in some community settings
- Health policy issues surrounding access, cost, and reimbursement

Call to Action

Develop Consensus Terminology and Messaging in Patient Education Materials

- Lack of consistent language to describe biomarker testing among patient advocacy organizations and industry is a barrier
 - To address this issue, advocacy groups and industry need to align on common terminology and messaging with regard to biomarkers (**Figure 2**)

Figure 2. Strategies for Developing Consistent Messaging in Patient-Facing Education Materials

Common Messaging; Different Wrappers

- Educational content developed in collaboration across organizations
 - Common content packaged in each organization's unique branding
- Common factsheets, brochures, etc. with the logos of all collaborators included at the bottom

One Voice; Individual Identity

- Collaboration to develop a shared consensus statement including:
 - Best practices
 - Common core items or 'building blocks'
- Each organization utilizes the consensus statement as a guide to develop their own materials

Increasing Patient Awareness

- Patient advocacy groups are well-positioned to address the lack of patient awareness with education campaigns
- For patient-facing education materials to be successful, they should conform to the 5 Cs of patient-facing materials (**Table 1**)

Table 1. The 5 Cs of Patient-Facing Education Materials

Consistent
• A cohesive message across lung cancer advocacy groups • Consistent terminology for the process itself (eg, biomarker testing vs genetic testing or molecular diagnostics) • Unified core key messages
Clear
• Simple messages and infographics • Easy-to-understand definitions • Sixth-grade literacy level (including proper introduction and explanation of multisyllable words)
Customizable
• Different levels of information available depending on patient and caregiver needs and preference – Basic information as a starting point – Additional resources available for patients who desire more information • Multiple languages available
Comprehensive
• Ensure that all materials are accurate, and that no information is missing • Ensure all relevant topics (Who, What, When, Where, Why, and How) are covered • Regularly review materials both internally and externally (by experts in the field) • Update materials regularly to keep pace with the constantly changing treatment landscape • Remove outdated materials from circulation
Checklists
• Key questions for patients to ask their physicians regarding biomarker testing

The Who, What, When, Where, Why, and How of Biomarker Testing

- As a first step, consistent messaging with regard to the Who, What, When, Where, Why, and How of biomarker testing are needed (**Table 2**)

Table 2. Finding a Unified Voice: The Who, What, When, Where, Why, and How of Biomarker Testing

WHO should get tested?
• All patients with advanced or recurrent NSCLC • For patients with early-stage disease, testing could be beneficial for inclusion in certain clinical trials • As additional targeted therapies become available, biomarker testing for patients with SCLC may also be recommended
WHAT is biomarker testing?
• Lung cancer tumors can grow and spread in different ways – Biomarker testing identifies changes that may define your unique cancer – Biomarker testing is the first step in precision medicine, whereby your treatment is matched to your specific tumor
WHEN should a patient get tested?
• At diagnosis, progression, and recurrence/relapse • If biomarker testing is not done at diagnosis, it should be done as soon as possible following diagnosis and prior to treatment
WHERE is testing done?
• When possible, biopsy should be done by a dedicated thoracic physician at a facility that does many biopsies per week • Biomarker testing may be done in-house or sent out to a testing facility
WHY is biomarker testing important?
• The effectiveness of different treatments varies greatly depending on each patient's biomarker profile • Patients with tumors that express certain markers may not respond as well to standard chemotherapy • Patients matched with the appropriate treatment based on biomarker testing may live a better and longer life than those who receive standard chemotherapy treatment
HOW is testing done?
• At the time of diagnosis, the doctor should send the patient's tissue, fluid, or blood to a lab for appropriate testing – The doctor may wait for the results (~7–14 days) before starting treatment in order to identify the best treatment for the patient

Initiatives Geared Toward Physicians

- Understanding gaps in physician knowledge regarding biomarker testing and developing initiatives to address these gaps are warranted
- The roundtable recommended a comprehensive and well-developed physician survey to gather information

Initiatives Geared Toward Physicians (continued)

- Results of the survey can then be used to identify gaps in physicians' knowledge and guide the development of educational initiatives to address gaps (**Figure 3**)
- Examples of information to be collected through the survey include:
 - Is tissue being collected for biomarker testing during the biopsy? Why or why not?
 - If samples are being collected, is it being done correctly? If incorrectly, what are the contributing factors?
 - Is a lung biomarker panel test (comprehensive profiling) being ordered? Why or why not?
 - Are physicians comfortable recommending treatment based on the results of biomarker testing? Why or why not?

Figure 3. Potential Physician-Geared Initiatives

Education via online forums
- Up-to-date
- Mobile applications

Physician checklists
- Key steps in the biomarker testing process
- Role of each member of the care team

Biomarker testing as a quality metric
- Percent of patients tested
- Percent not tested
- Explanation for why not tested

Conclusions

- **Additional education and awareness are needed to establish biomarker testing as part of standard of care in patients with advanced-stage lung cancer**
- **The ultimate goal is that every patient has a full biomarker panel available (or at least tests in progress) at their first appointment with an oncologist**
- **A unified effort from lung cancer advocacy organizations, healthcare providers, and industry partners is needed to achieve this goal**

References

1. Kris MG, et al. *JAMA*. 2014;311:1998–2006.
2. Mok TS, et al. *N Engl J Med*. 2009;361:947–57.
3. Peters S, et al. *N Engl J Med*. 2017;377:829–38.
4. Solomon BJ, et al. *N Engl J Med*. 2014;371:2167–77.

Disclosures

WB, CA, UBR, AC, DD, HG, CL, LSR, CS, GAS, RAS, and LW have no conflicts to disclose.

Acknowledgements

All listed authors meet the criteria for authorship set forth by the International Committee for Medical Journal Editors. Editorial support (assembling tables and figures, collating author comments, copyediting, fact checking, referencing, and graphic services) were provided by AOIC, LLC, and were funded by AbbVie, Inc.

Presented at the International Association for the Study of Lung Cancer 19th World Congress on Lung Cancer · Toronto, Canada (September 23–26, 2018)